218

Safety and Efficacy of Anti-PD-1 Antibody Dostarlimab in Patients with Mismatch Repair-Deficient (dMMR) GI Cancers

Thierry André,[1] Dominique Berton,[2] Filippo De Braud,[3] Giuseppe Curigliano,[4] Wei Guo,[5] Hadi Danaee,[5] Sharon Lu,[5] Ellie Im,[5] and Naureen Starling[6]

[1]Sorbonne Université, Medical Oncology Department, Hôpital Saint-Antoine, Paris, France; [2]Service de Cancérologie Médicale, ICO Centre René Gauducheau, Saint-Herblain, France; [3]Medical Oncology and Hematology, Fondazione IRCCS Istituto Nazionale dei Tumori and Università degli Studi Milan, Milan, Italy; [4]Department of Oncology and Hemato-Oncology, University of Milan, Milan, Italy; [5]GlaxoSmithKline, Waltham, MA, USA; [6]GI Cancer Unit, The Royal Marsden Hospital, London, England

Background

- Dostarlimab (TSR-042) is an investigational, humanized, anti-programmed death (PD)-1 monoclonal antibody that binds the PD-1 receptor, blocking the interaction with ligands PD-L1 and PD-L2[1]
- The ongoing GARNET trial (NCT02715284) is evaluating dostarlimab in patients with advanced solid tumors
- Here, we present safety and efficacy data from patients with mismatch repair-deficient (dMMR) non-endometrial cancers, including gastrointestinal (GI)

Objectives

- The primary objective was to measure antitumor activity by objective response rate (ORR) and duration of response (DOR) assessed by blinded independent central review per Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST v1.1)

Methods

- This cohort from the GARNET trial enrolled patients with dMMR or microsatellite instability-high (MSI-H) non-endometrial solid tumors (**Figure 1**), the majority of which were GI in origin
- Patients must have demonstrated disease progression following prior systemic therapy for advanced disease, as assessed by blinded independent central review
- Patients received 500 mg of dostarlimab every 3 weeks (Q3W) for 4 cycles and 1000 mg every 6 weeks (Q6W) thereafter for up to 2 years or until disease progression or discontinuation
- Patients were included in the efficacy analysis if they received ≥1 dose of dostarlimab, had measurable disease at baseline, and had ≥24 weeks of follow-up by the data cutoff date, July 8, 2019
- All patients who received ≥1 dose of dostarlimab were included in the safety analysis

Figure 1. GARNET Trial Design

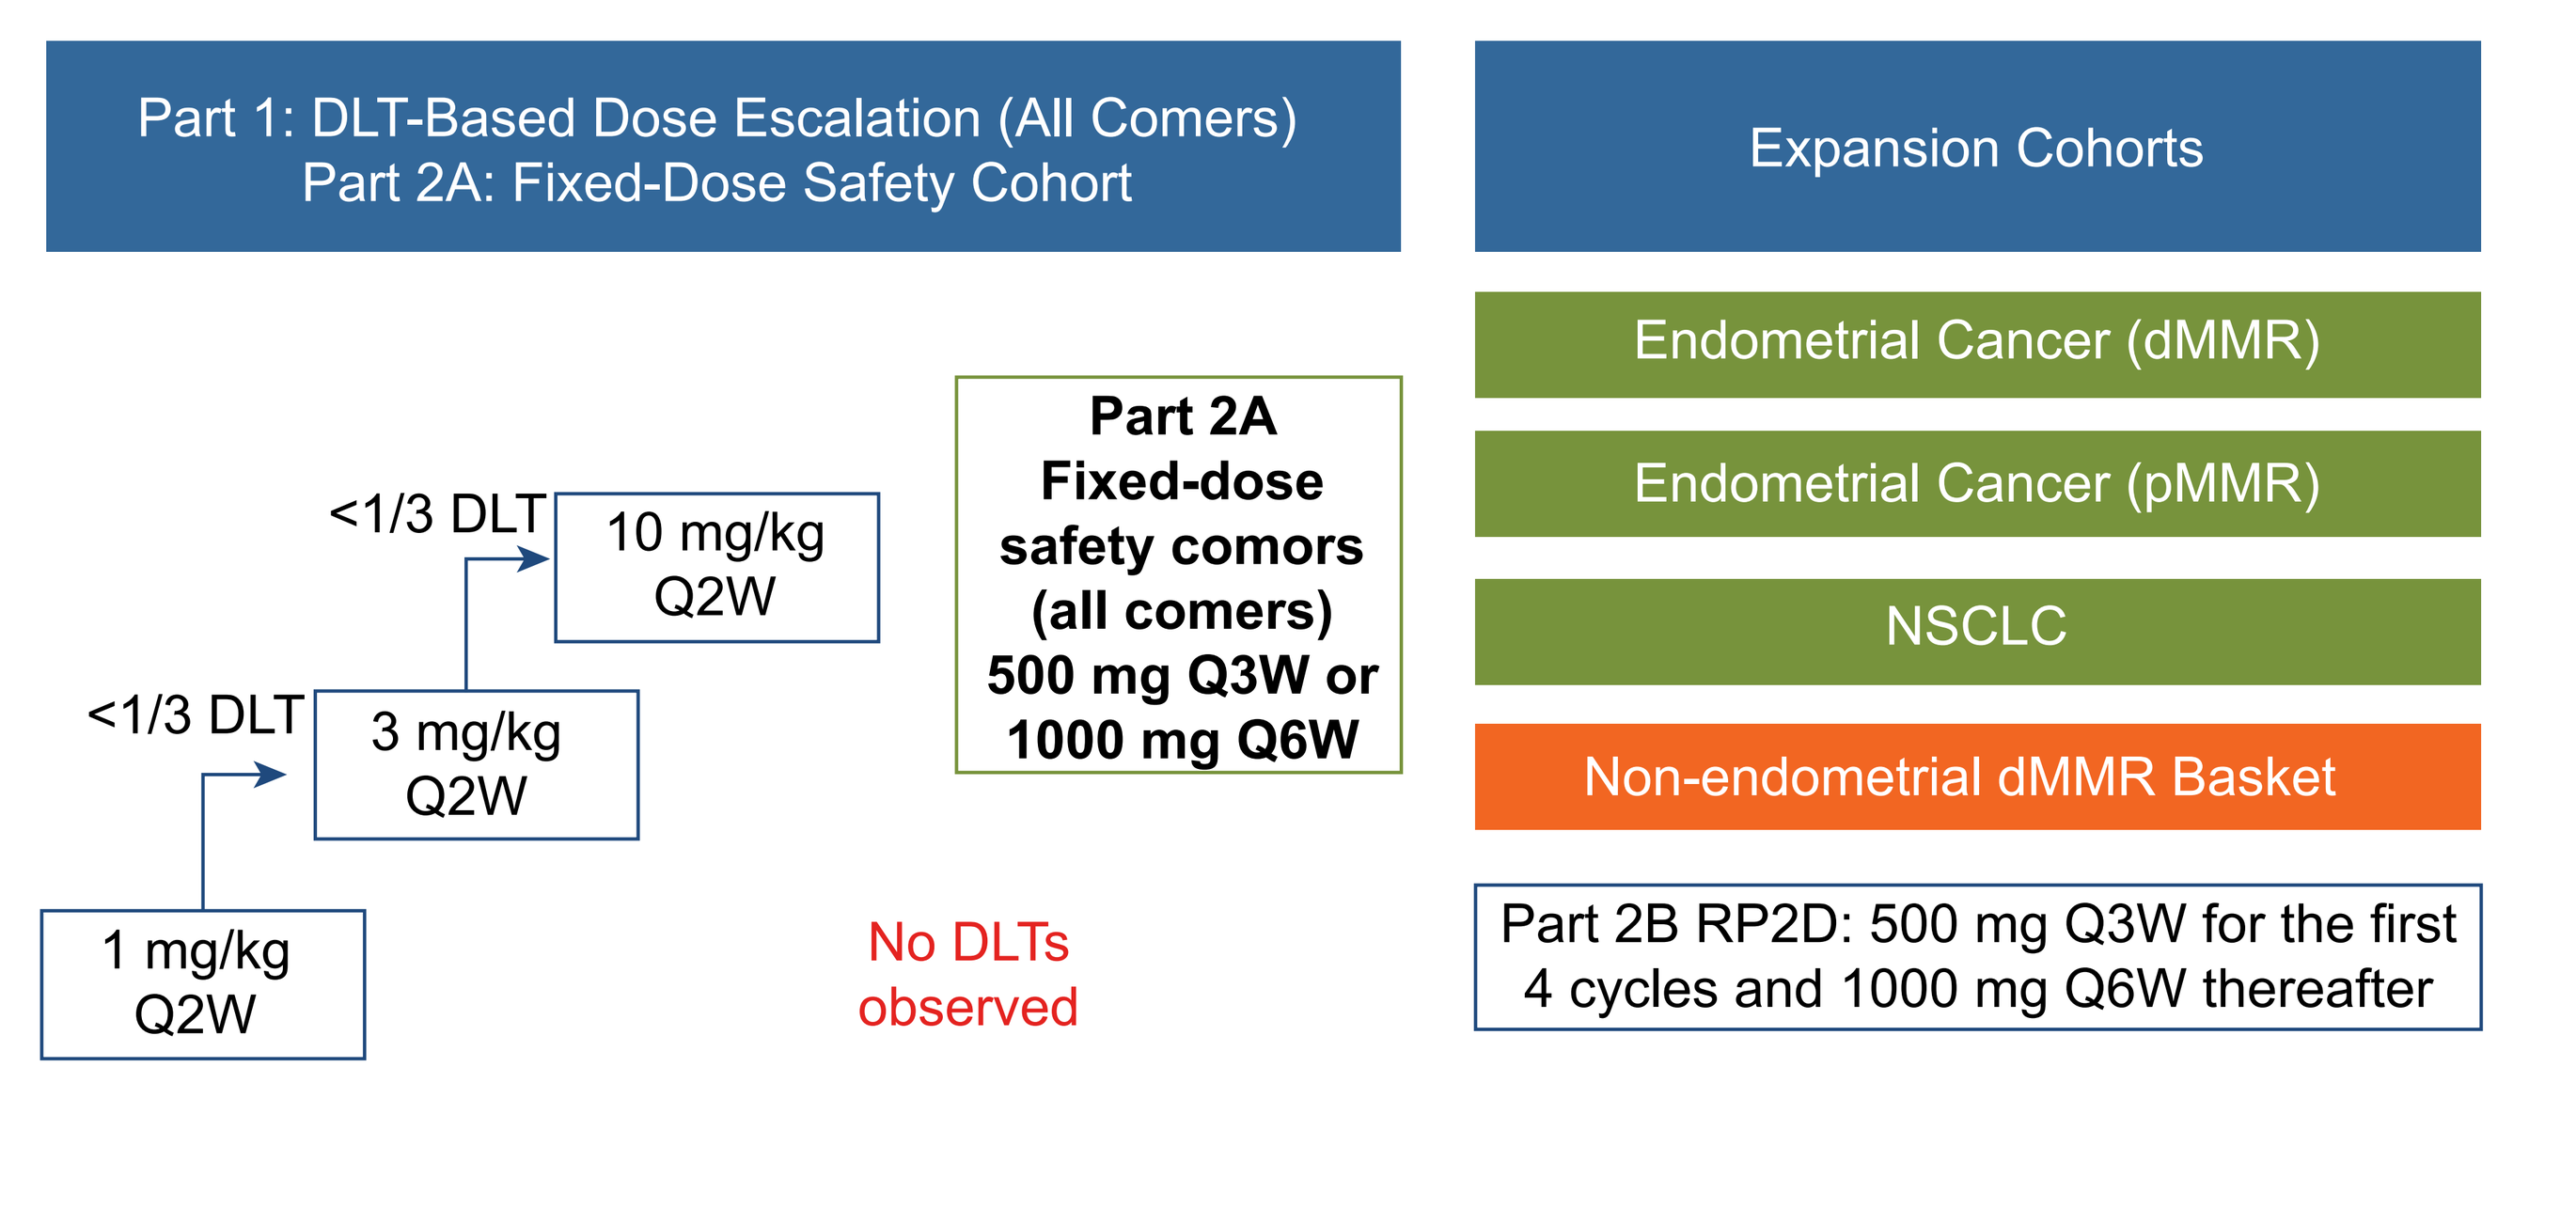
Part 1: DLT-Based Dose Escalation (All Comers)
Part 2A: Fixed-Dose Safety Cohort
Expansion Cohorts
Endometrial Cancer (dMMR)
Endometrial Cancer (pMMR)
NSCLC
Non-endometrial dMMR Basket
Part 2A Fixed-dose safety comors (all comers) 500 mg Q3W or 1000 mg Q6W
<1/3 DLT
10 mg/kg Q2W
<1/3 DLT
3 mg/kg Q2W
1 mg/kg Q2W
No DLTs observed
Part 2B RP2D: 500 mg Q3W for the first 4 cycles and 1000 mg Q6W thereafter

DLT=dose-limiting toxicity; dMMR=mismatch repair-deficient; NSCLC=non-small cell lung cancer; pMMR=mismatch repair-proficient; Q2W=every 2 weeks; Q3W=every 3 weeks; Q6W=every 6 weeks; RP2D=recommended phase 2 dose.

Results

- The efficacy analysis included 48 patients as of the data cutoff date (July 8, 2019)
- Of the efficacy population, 42 (88%) patients had GI tumors (**Table 1**), including 30 with colorectal cancer

Table 1. Patient Demographics and Baseline Characteristics, Efficacy Population

Characteristic	Cohort F N=48
Median age, years (range)	64.0 (24–80)
Sex, n (%)	
Male	24 (50)
Female	24 (50)
ECOG performance status, n (%)	
0	18 (37)
1	30 (63)
Histology, n (%)	
Colorectal	30 (63)
Small intestine	8 (17)
Gastric	4 (8)
Ovarian	2 (4)
Other[a]	4 (8)
Disease stage at first diagnosis, n (%)	
I	2 (4)
II	6 (13)
III	15 (31)
IV	24 (50)
Unknown	1 (2)
Number of prior lines of therapy for advanced disease, n (%)	
0	4 (8)
1	16 (33)
2	23 (48)
3	4 (8)
≥4	1 (2)
Categories of prior therapy, n (%)	
Surgery	44 (92)
Radiotherapy	13 (27)
Bevacizumab	13 (27)

[a]Adrenal cortical, genital neoplasm malignant female, renal cell, and unknown.
ECOG=Eastern Cooperative Oncology Group.

Efficacy

- Confirmed ORR in patients with dMMR was 44% (95% CI, 29.5–58.8), with a complete response rate of 8%
- ORR was consistent across both colorectal and non-colorectal tumor types (**Table 2**)
- With a median follow-up of 8.1 months, median DOR was not reached (range: 2.8–13.8 months)
- The probability of maintaining response at 12 months was 86%

Table 2. Objective Response Rate by Tumor Type

Tumor type	Patients, N	Confirmed ORR (RECIST v1.1) n (%)	Confirmed ORR (RECIST v1.1) 95% CI
Overall	**48**	**21 (44)**	**(29.5–58.8)**
CRC	**30**	**13 (43)**	**(25.5–62.6)**
Non-CRC	**18**	**8 (44)**	**(21.5–69.2)**
Small intestine	8	2 (25)	(3.2–65.1)
Gastric	4	2 (50)	(6.8–93.2)
Ovarian	2	PR, SD	
Adrenal cortical	1	PR	
Genital neoplasm malignant female	1	PR	
Renal cell	1	SD	
Unknown	1	PR	

CRC=colorectal cancer; ORR=objective response rate; PR=partial response; RECIST=Response Evaluation Criteria in Solid Tumors; SD=stable disease.

- Change in tumor volume and duration of response are shown in **Figure 2** and **Figure 3**, respectively

Figure 2. Best Response by Tumor Volume Measurement

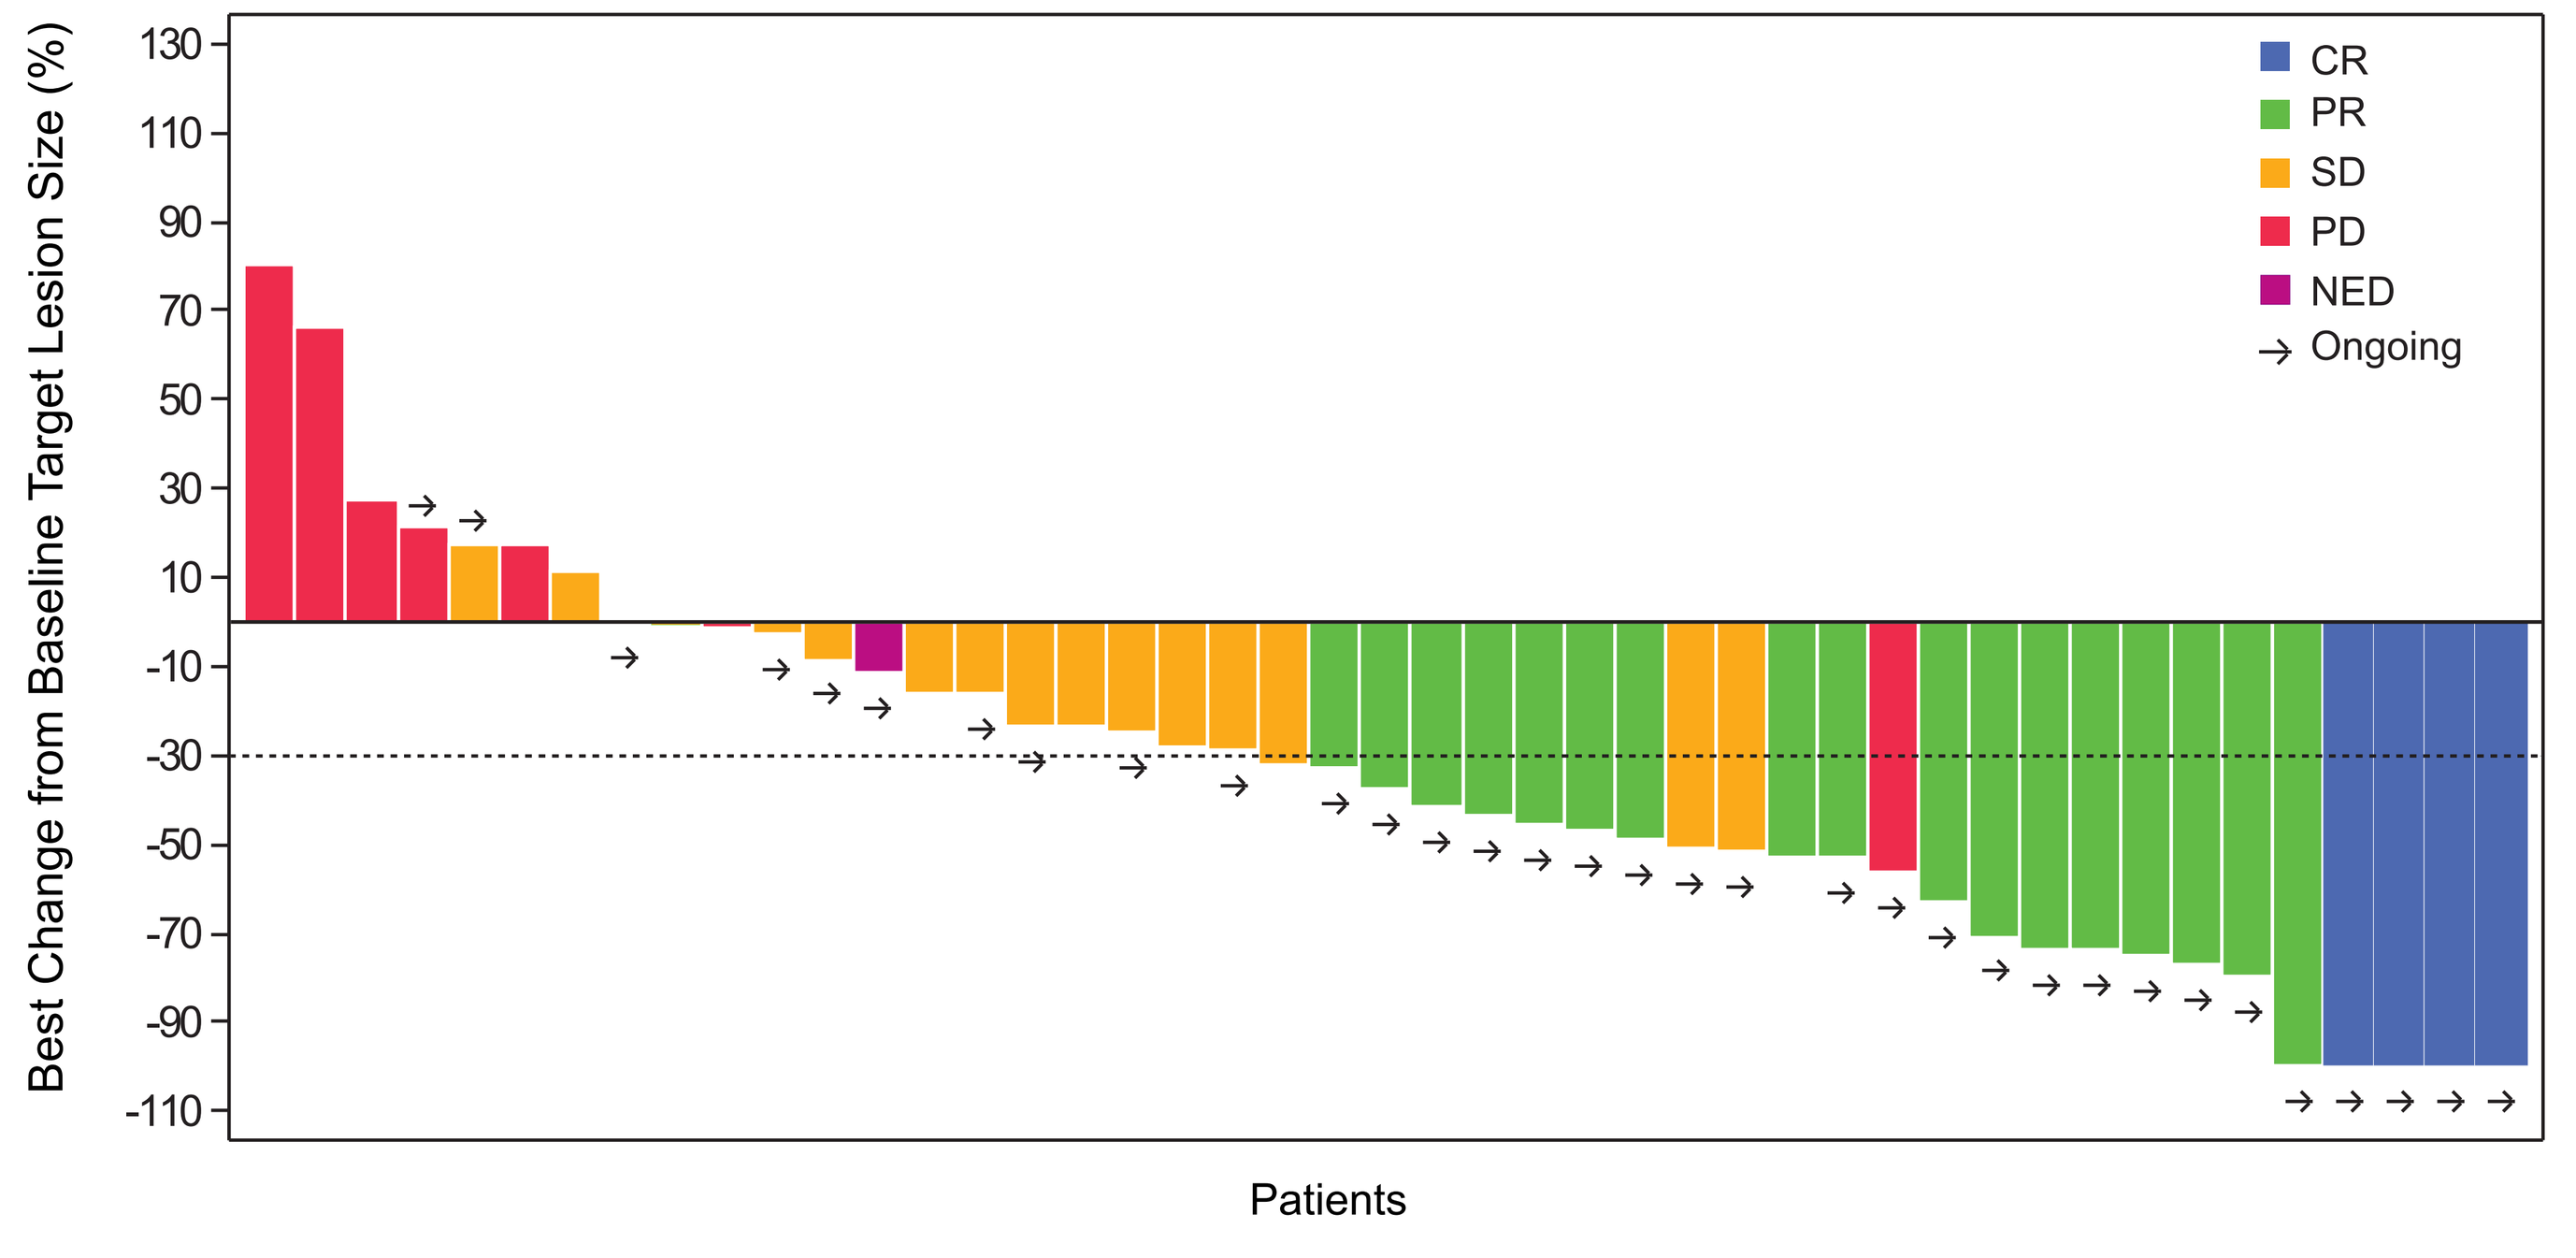
Best Change from Baseline Target Lesion Size (%)
130
110
90
70
50
30
10
-10
-30
-50
-70
-90
-110
Patients
CR
PR
SD
PD
NED
Ongoing

CR=complete response; NED=no evidence of disease; PD=progressive disease; PR=partial response; SD=stable disease.

Figure 3. Duration of Response

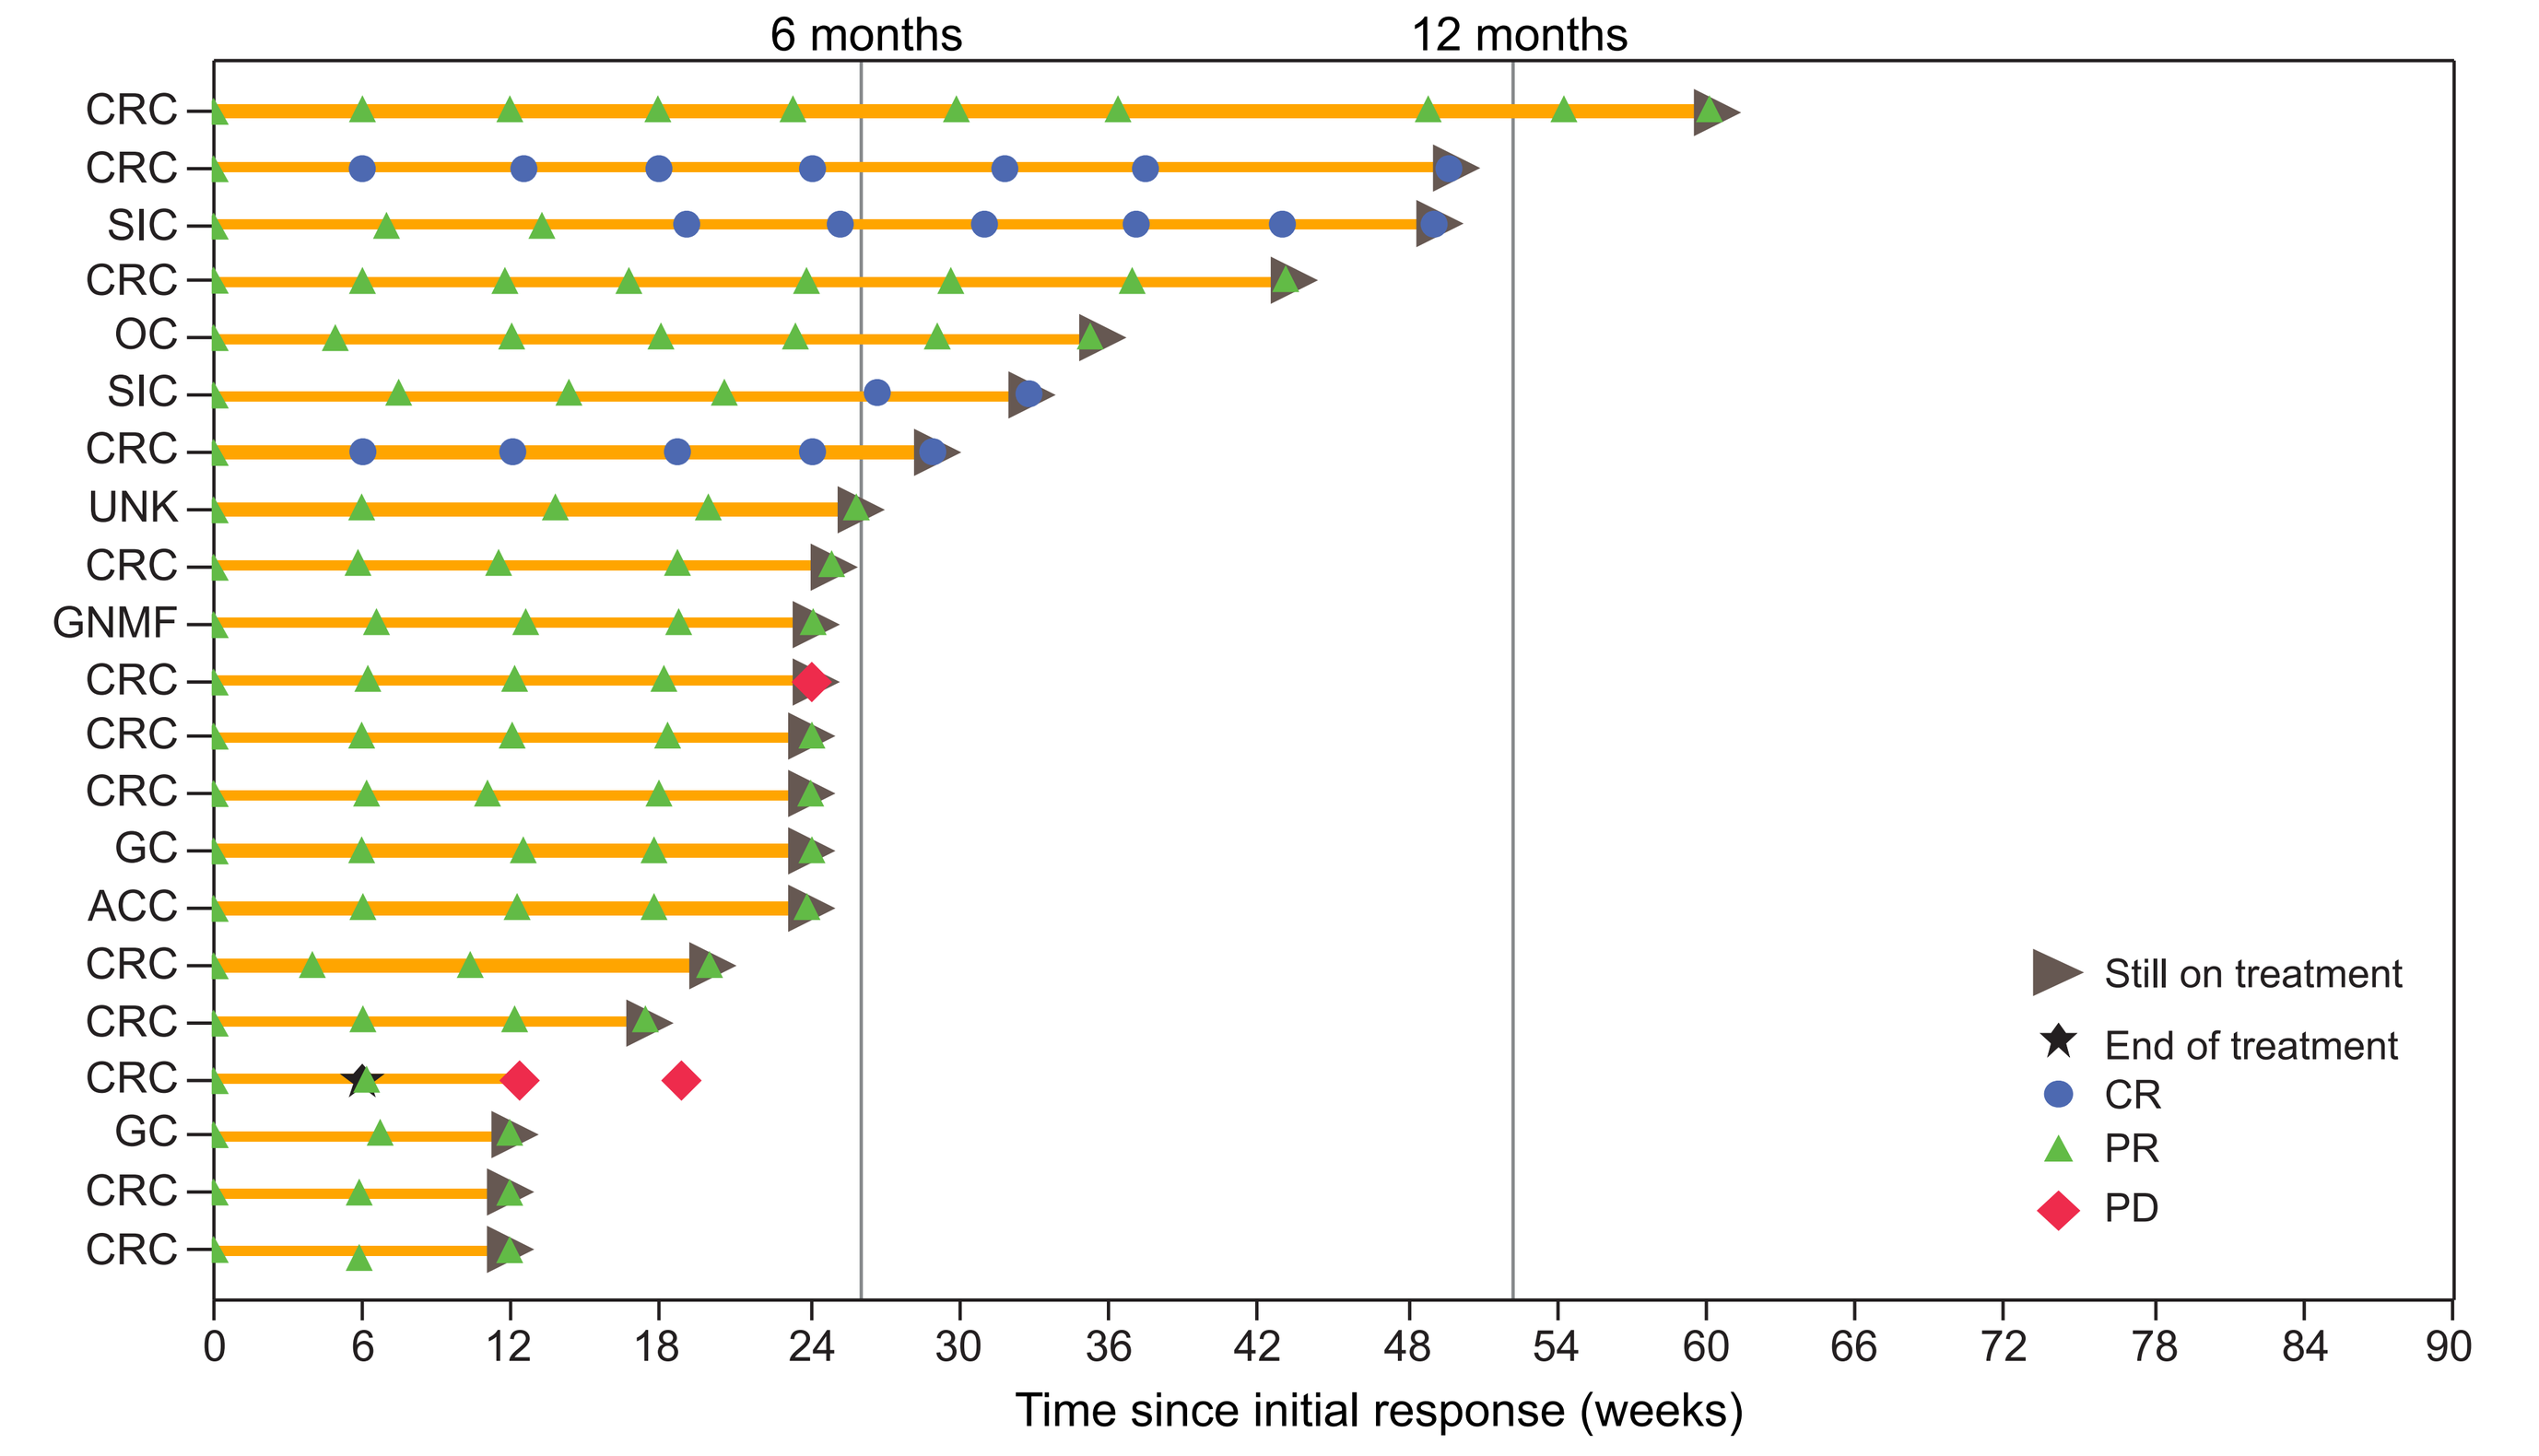
6 months
12 months
CRC
CRC
SIC
CRC
OC
SIC
CRC
UNK
CRC
GNMF
CRC
CRC
CRC
GC
ACC
CRC
CRC
CRC
GC
CRC
CRC
0
6
12
18
24
30
36
42
48
54
60
66
72
78
84
90
Time since initial response (weeks)
Still on treatment
End of treatment
CR
PR
PD

ACC=adrenal cortical cancer; CR=complete response; CRC=colorectal cancer; GC=gastric cancer; GNMF=genital neoplasm malignant female; OC=ovarian cancer; PD=progressive disease; PR=partial response; SIC=small intestine cancer; UNK=unknown.

Safety

- The safety population included 109 patients who received ≥1 dose of dostarlimab
- Discontinuations due to treatment-related adverse events (TRAEs) were reported in 2 (2%) patients (hypersensitivity and pneumonitis)
- Two deaths due to treatment-emergent adverse events were reported (cerebrovascular accident and pneumonia); none were assessed as related to dostarlimab
- Any grade TRAEs were reported in 60 (55%) patients
- Grade ≥3 TRAEs were reported in 8 (7%) patients
- Any grade and grade ≥3 TRAEs are shown in **Table 3**
- Grade ≥2 immune-related TRAEs occurred in 20 (18%) patients; 5 (5%) were grade ≥3 (**Table 4**)

Table 3. Any Grade and Grade ≥3 TRAEs

Preferred term, n (%)	Cohort F N=109
Any grade TRAEs occurring in ≥5% of patients	
Asthenia	14 (13)
Pruritus	11 (10)
Rash	11 (10)
Diarrhea	10 (9)
Arthralgia	9 (8)
ALT increased	7 (6)
AST increased	7 (6)
Hypothyroidism	7 (6)
Grade ≥3 TRAEs	
Rash	2 (2)
Adrenal insufficiency	1 (1)
Anemia	1 (1)
Diarrhea	1 (1)
Fatigue	1 (1)
Hyperlipasemia	1 (1)
Hyperthyroidism	1 (1)
Lower respiratory tract infection	1 (1)
Neutropenia	1 (1)

ALT=alanine aminotransferase; AST=aspartate aminotransferase; TRAE=treatment-related adverse event.

Table 4. Immune-Related TRAEs

Preferred term, n (%)	Cohort F N=109
Grade ≥2 irTRAEs occurring in >1 patient	
ALT increased	3 (3)
Arthralgia	3 (3)
Diarrhea	3 (3)
Hypothyroidism	3 (3)
Rash	3 (3)
Adrenal insufficiency	2 (2)
Hyperthyroidism	2 (2)
Pneumonitis	2 (2)
Grade ≥3 irTRAEs	
Rash	2 (2)
Adrenal insufficiency	1 (1)
Diarrhea	1 (1)
Hyperthyroidism	1 (1)

ALT=alanine aminotransferase; irTRAE=immune-related treatment-related adverse event.

Conclusions

- Dostarlimab demonstrated robust, durable antitumor activity in a cohort of patients with dMMR non-endometrial solid tumors, the majority of which were GI cancers
- Most TRAEs were low grade; immune-related TRAEs were rare and low grade
- Adverse events were characteristic of anti-PD-1 therapies

REFERENCES

1. Laken H, et al. *Eur J Cancer.* 2016;69(Suppl 1):S102.

ACKNOWLEDGEMENTS

The authors thank the patients and their families for their participation in this study, as well as the study teams at each of the study sites. Writing and editorial support, funded by GlaxoSmithKline (Waltham, MA, USA) and coordinated by Heather Ostendorff-Bach, PhD, of GSK, were provided by Eric Scocchera, PhD, and Anne Cooper, MA, of Ashfield Healthcare Communications (Middletown, CT, USA).

thierry.andre@aphp.fr

Presented at the 2020 Gastrointestinal Cancers Symposium Annual Meeting | January 23-25, 2020 | San Francisco, CA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.